An evaluation of effectiveness of high-concentration capsaicin topical system (HCCTS) across RCT vs RWE data for 1 year treating painful diabetic peripheral neuropathy

Samuel Allen, PhD[1]; Audrey Carnevale, PhD[1]; Lizandra Marcondes, MD, PhD[1]; Mariëlle Eerdekens, MD[2]

1. Averitas Pharma, Morristown, NJ, USA 07960, 2. Grünenthal GmbH, Aachen, Germany.

Purpose

We sought to provide a side-by-side comparison of a RCT (PACE) and RWE (CASPAR), focusing on differences and similarities in design and results for 1 year of treatment with HCCTS for pDPN.

Background

- Painful diabetic peripheral neuropathy (pDPN) is a chronic complication of diabetes associated with a negative impact on quality of life.[1,2]
- In pDPN, chronic hyperglycemia causes damage to peripheral nerves,[3] accelerated by poor microcirculation.[4]
- The pain in pDPN is chronic and requires ongoing treatment to achieve long-term analgesia. If left untreated, progression of DPN can lead to loss of normal protective sensation, ulcers, infections, and, in severe cases, amputation.[5]
- Metabolic control alone does not prevent development and progression of pDPN and all patients with diabetes should have regular foot health checks and, if diagnosed, appropriate treatment should be started in a timely fashion.[4]
- High-concentration capsaicin 8% topical system (HCCTS; Figure 1) is approved for the treatment of neuropathic pain associated with DPN of the feet and post-herpetic neuralgia.[1]
- PACE was a randomized controlled trial (RCT) of HCCTS + standard of care (SOC) vs SOC alone. PACE demonstrated that repeated HCCTS treatment was not associated with adverse effects on neurological function and provided evidence (secondary outcomes) for efficacy over 1 year.[6,7]
- Recently, Überall et al. reported on data from CASPAR, a 1-year observational study of HCCTS.[8]

Figure 1: Application of the capsaicin 8% topical system in the office of the healthcare professional

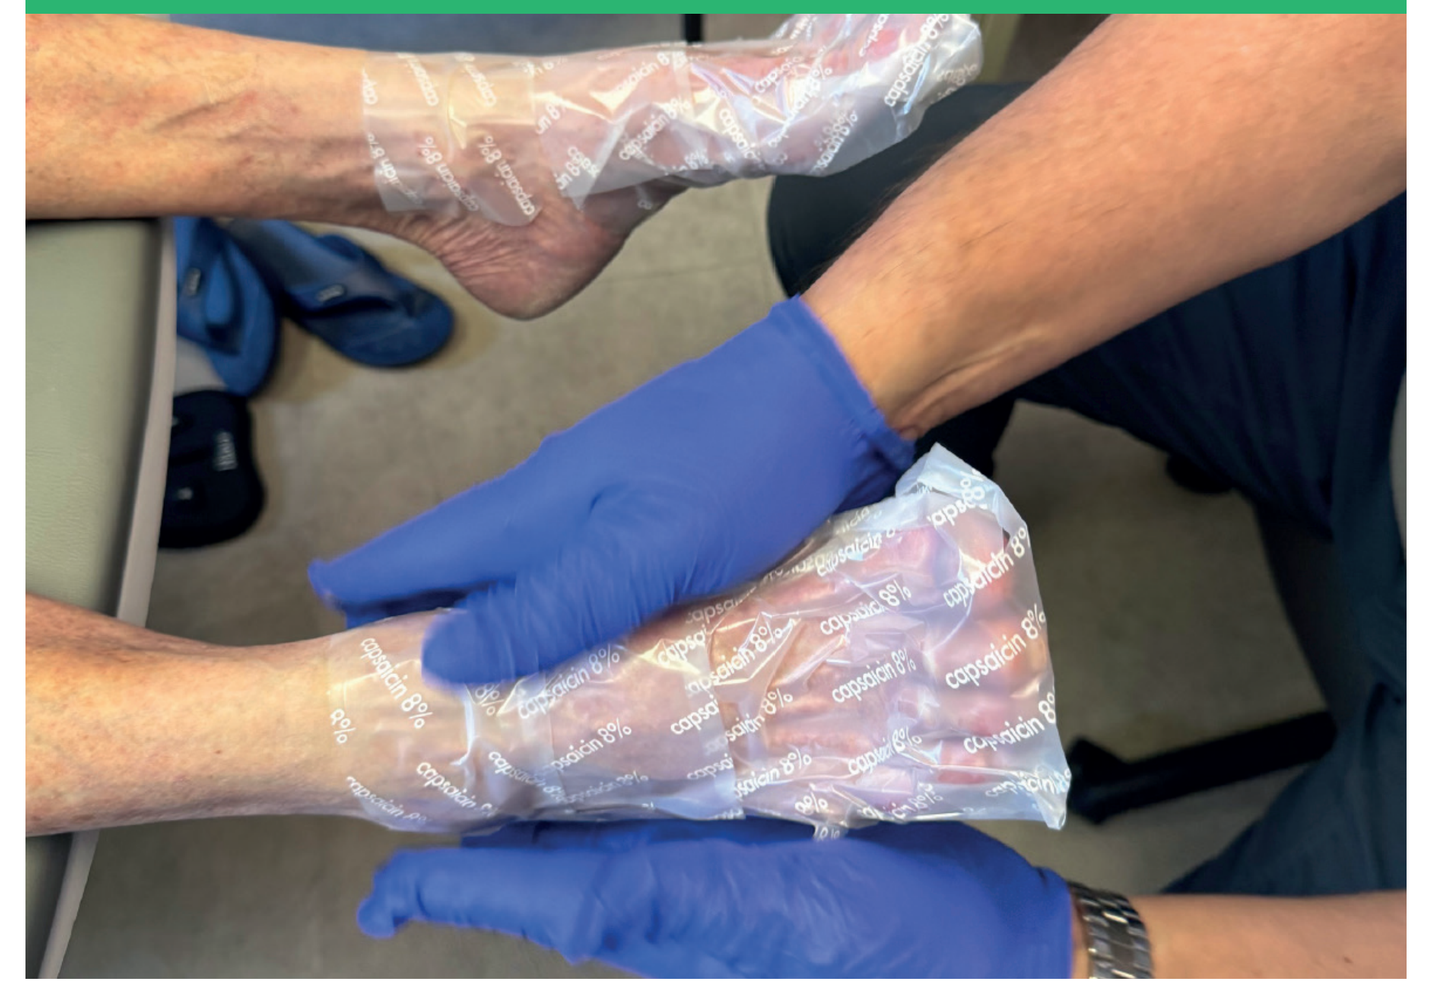

Figure used with permission from Dr. Gary Graf, APRN

- Evidence provided by RCTs can be complemented by real-world evidence (RWE) from observational studies.[9,10]
 - RCTs evaluate the efficacy and safety of treatments in a highly controlled and well-balanced group of patients. Patients in RCTs represent only a small fraction of patients who may require treatment in the real world.
 - While the real-world applicability of observational data can sometimes be greater than that of RCTs, observational studies may have more biases and confounding factors compared with the more controlled environment of RCTs.

Interpretation

- Comparing data from PACE and CASPAR is challenging due to differences in the instruments used for outcome collection, the graphical representation of the results, and the study designs.
- Compared with PACE, CASPAR included a patient population that was, on average, older; duration of pDPN and pain intensity at baseline were similar in both studies.
- In both studies, each successive treatment was associated with a decrease in average pain intensity and sleep interference, as well as an increase in responder rate.
- In CASPAR:
 - The greatest benefit was observed in patients who received 4 HCCTS treatments.
 - When patients stopped taking HCCTS, the benefit stopped, and the pain intensity and sleep interference trended back toward baseline levels.
- The magnitude of changes (e.g., response rate) may appear larger in CASPAR compared with PACE. This may be due to confounding factors not controlled for in an observational study, in particular selection bias (i.e., healthcare professionals may have selected patients more likely to respond to HCCTS and excluded those less likely to).
- The difference in magnitude of changes might be reflective of both how RWE more easily generalizes to clinical practice than RCTs and how RCTs are restrictive, with multiple inclusion criteria.
- HCCTS was well tolerated in both studies. The proportion of patients reporting application-site reactions was higher in CASPAR than in PACE, but in CASPAR, this was lower for patients who received multiple treatments.

Trial Design

PACE (RCT)		CASPAR (RWE)
Prospective, randomized controlled trial	DESIGN	Retrospective, single-arm observational study*
1 YEAR	DURATION	1 YEAR
71 sites in 11 European countries	LOCATIONS	283 sites in Germany
up to 7 HCCTS TREATMENTS applied for 30 min to feet + SOC	STUDY TREATMENT (IN THIS ANALYSIS)	up to 4 HCCTS TREATMENTS applied for 30 min to feet + SOC
Study visits were at months 0 (baseline), 2, 4, 6, 8, and 12. HCCTS could be re-applied when required at scheduled and/or unscheduled visits, provided treatment interval was ≥8 weeks (≥8 WEEKS)	HOW FREQUENTLY WAS IT RE-APPLIED?	When required, provided treatment interval was >12 weeks (>12 WEEKS)
SOC ALONE	COMPARATOR	NONE
% change from baseline in Norfolk QoL-DN total score (assesses impact of nerve dysfunction on QoL)	PRIMARY OUTCOME	% of patients who responded to HCCTS with repeated treatments
At baseline visit, monthly (alternate telephone visits and study visits), termination visit	WHEN DID PATIENTS ANSWER THE QUESTIONNAIRE?	At baseline (shortly before first treatment) and immediately prior to each treatment (for those who discontinued HCCTS, every 3 months)

*Data from German Pain eRegistry.

Baseline Characteristics, Mean

	PACE (RCT) HCCTS 30 min + SOC	PACE (RCT) SOC alone	CASPAR (RWE) HCCTS 30 min
Diagnosis (in this analysis)	pDPN of the feet	pDPN of the feet	pDPN of the feet
Number of patients	156	155	365
Mean age (years)	60.9	59.1	66.1
Mean BMI (kg/m²)	30.1	31.2	31.5
Mean duration of pDPN (years)	4.1	4.4	4.6

Pain Intensity

PACE (RCT)		CASPAR (RWE)
Item 5 of the Brief Pain Inventory (BPI), which asks the patient about average daily pain: 0 = no pain, 10 = pain as bad as you can imagine	HOW WAS IT MEASURED?	Visual analog scale (VAS), a unidimensional measure of average pain intensity in the preceding 24 hours: 0 = no pain, 100 = worst pain conceivable
5.6 (out of 10) · 5.5 (out of 10)	BASELINE VALUE	57.7 (out of 100)

By end of study, there was a 37.5% reduction from baseline in average daily pain with HCCTS 30 min + SOC

In the patients who received 4 HCCTS treatments, average daily pain score reduced from 58.4 to 14.2 at 12 months

Mean change from baseline in average daily pain

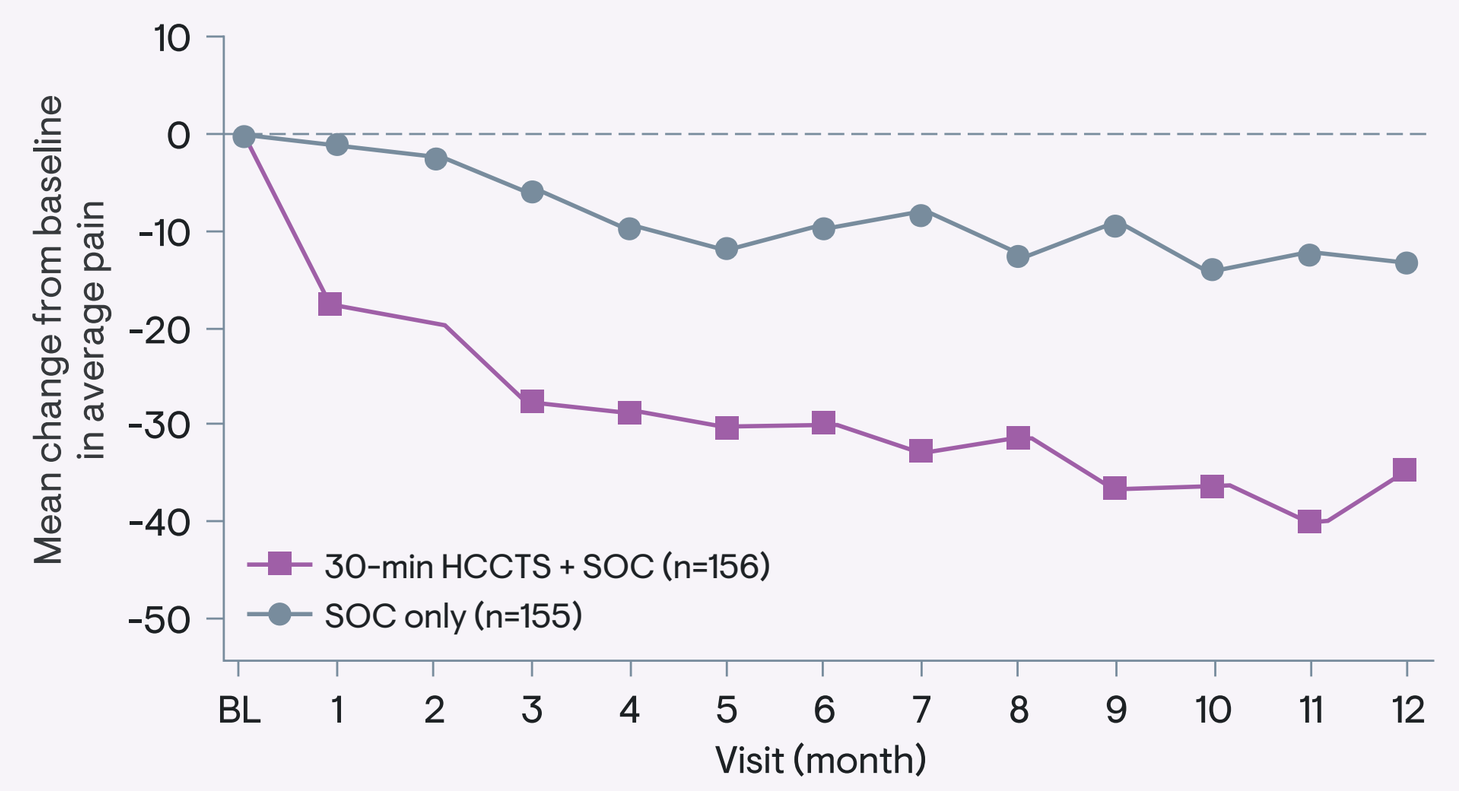
Mean change from baseline in average pain
10
0
-10
-20
-30
-40
-50
BL 1 2 3 4 5 6 7 8 9 10 11 12
Visit (month)
30-min HCCTS + SOC (n=156)
SOC only (n=155)

Mean score in average daily pain

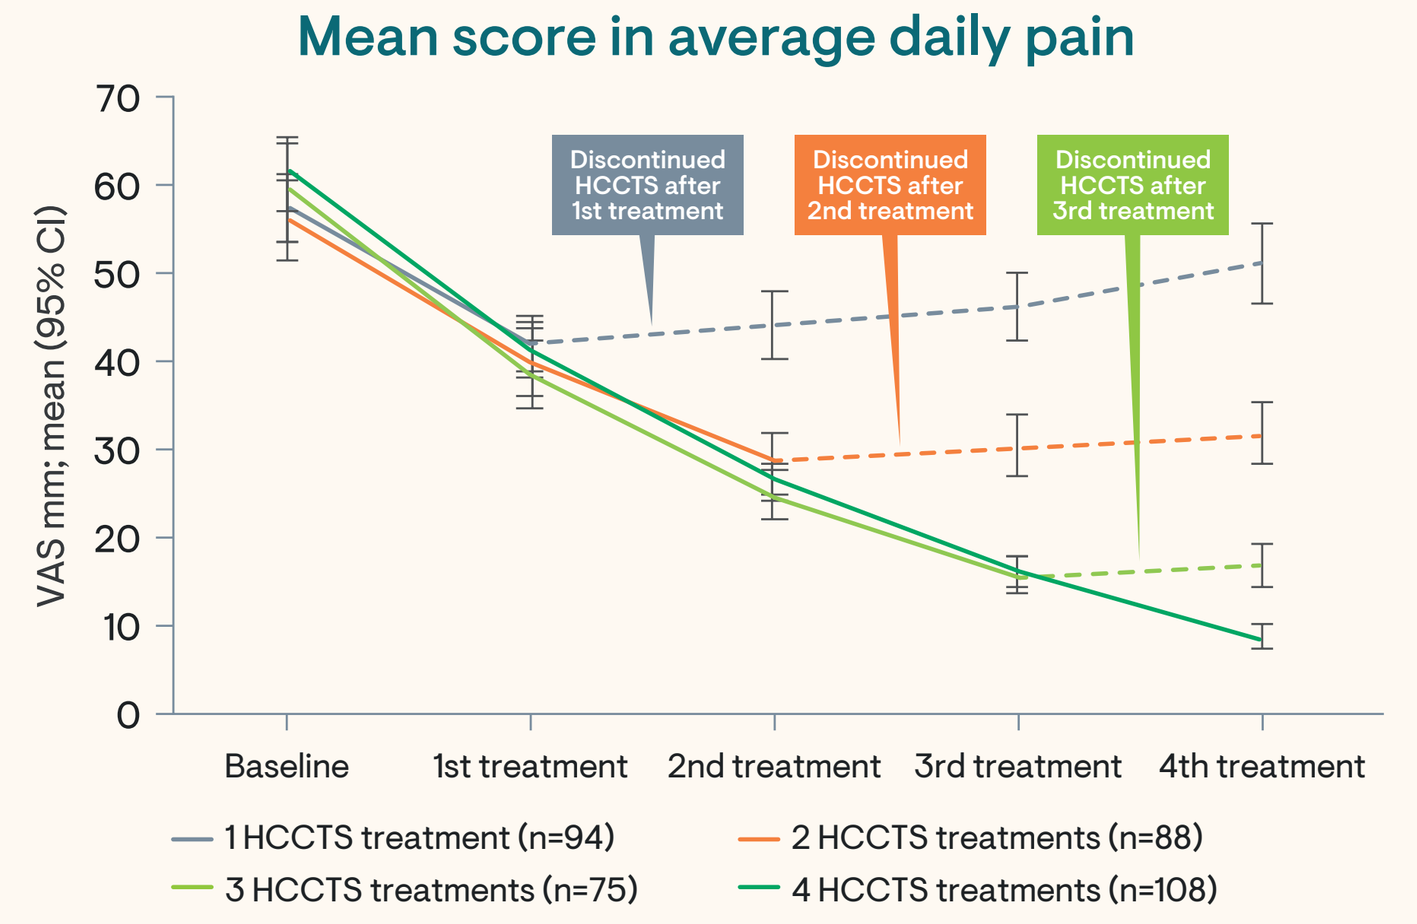
VAS mm; mean (95% CI)
70 60 50 40 30 20 10 0
Discontinued HCCTS after 1st treatment
Discontinued HCCTS after 2nd treatment
Discontinued HCCTS after 3rd treatment
Baseline
1st treatment
2nd treatment
3rd treatment
4th treatment
1 HCCTS treatment (n=94)
2 HCCTS treatments (n=88)
3 HCCTS treatments (n=75)
4 HCCTS treatments (n=108)

Responder Rate

Percentage of patients achieving ≥30% reduction in average daily pain in patients who received the maximum 7 HCCTS treatments[11]

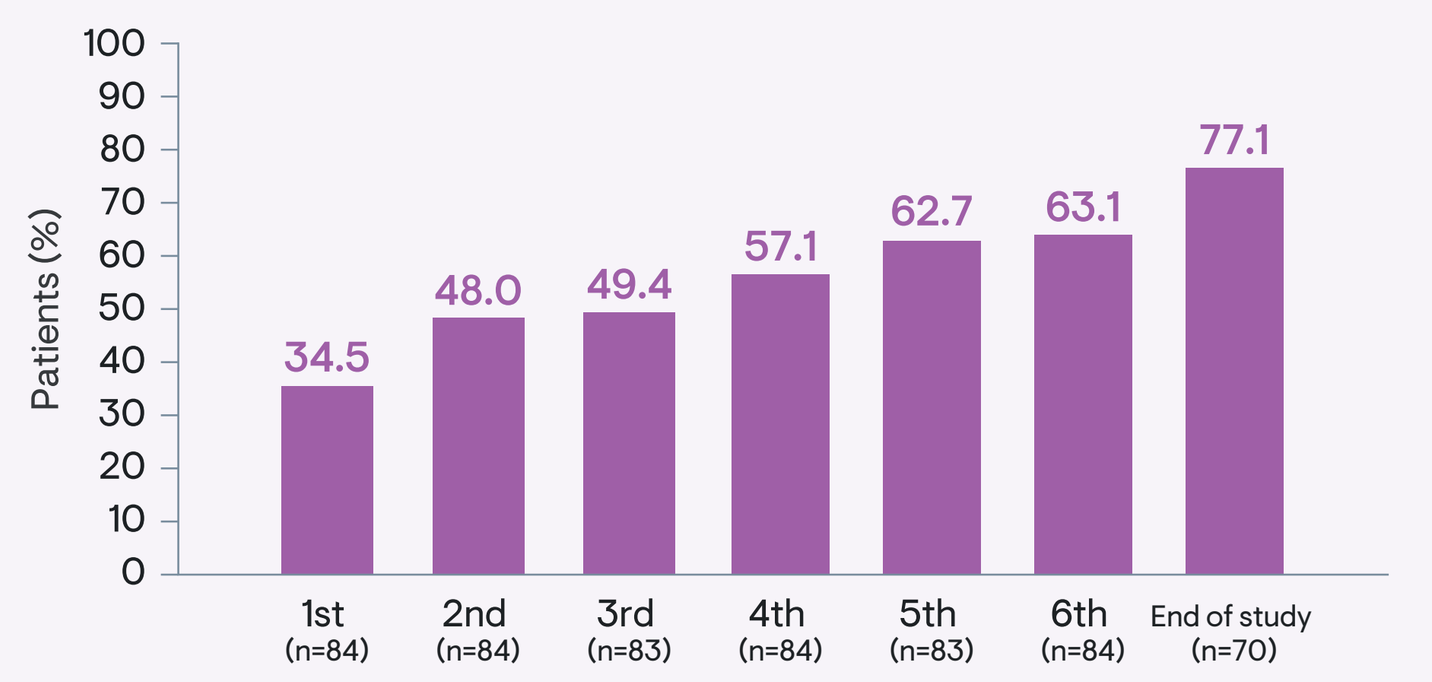
Patients (%)
100 90 80 70 60 50 40 30 20 10 0
34.5
48.0
49.4
57.1
62.7
63.1
77.1
1st (n=84)
2nd (n=84)
3rd (n=83)
4th (n=84)
5th (n=83)
6th (n=84)
End of study (n=70)

Percentage of patients achieving ≥30% reduction in average daily pain in patients who received all 4 treatments

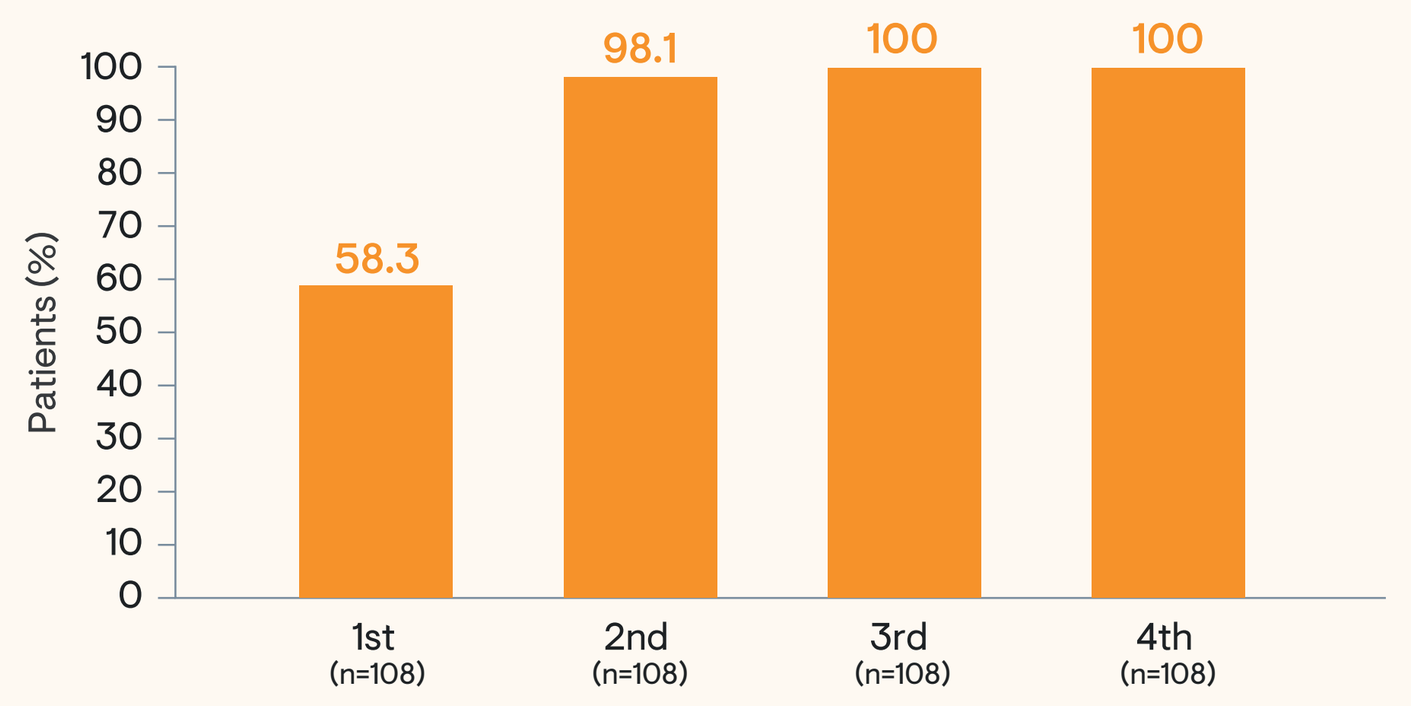
Patients (%)
100 90 80 70 60 50 40 30 20 10 0
58.3
98.1
100
100
1st (n=108)
2nd (n=108)
3rd (n=108)
4th (n=108)

Conclusion

Similar trends regarding effectiveness and tolerability, excluding application-site erythema, were observed after 1 year of HCCTS treatment for pDPN in both PACE (RCT) and CASPAR (RWE). The similarities corroborate the results from PACE, suggesting they can be generalized to a wider patient population, and demonstrating there is ongoing benefit of HCCTS treatment in real-world clinical practice.

Pain-Related Sleep Disturbance

PACE (RCT)		CASPAR (RWE)
BPI Diabetic Neuropathy (BPI-DN) Pain Interference Index: Question 9f. Patients rated the degree to which pain interfered with their sleep: 0 = does not interfere, 10 = completely interferes	HOW WAS IT MEASURED?	Modified Pain Disability Index (mPDI): Patients rated the degree to which pain affected their sleep: 0 = not at all, 100 = completely

BPI-DN Pain Interference Index: Q9f: Pain interfered with sleep

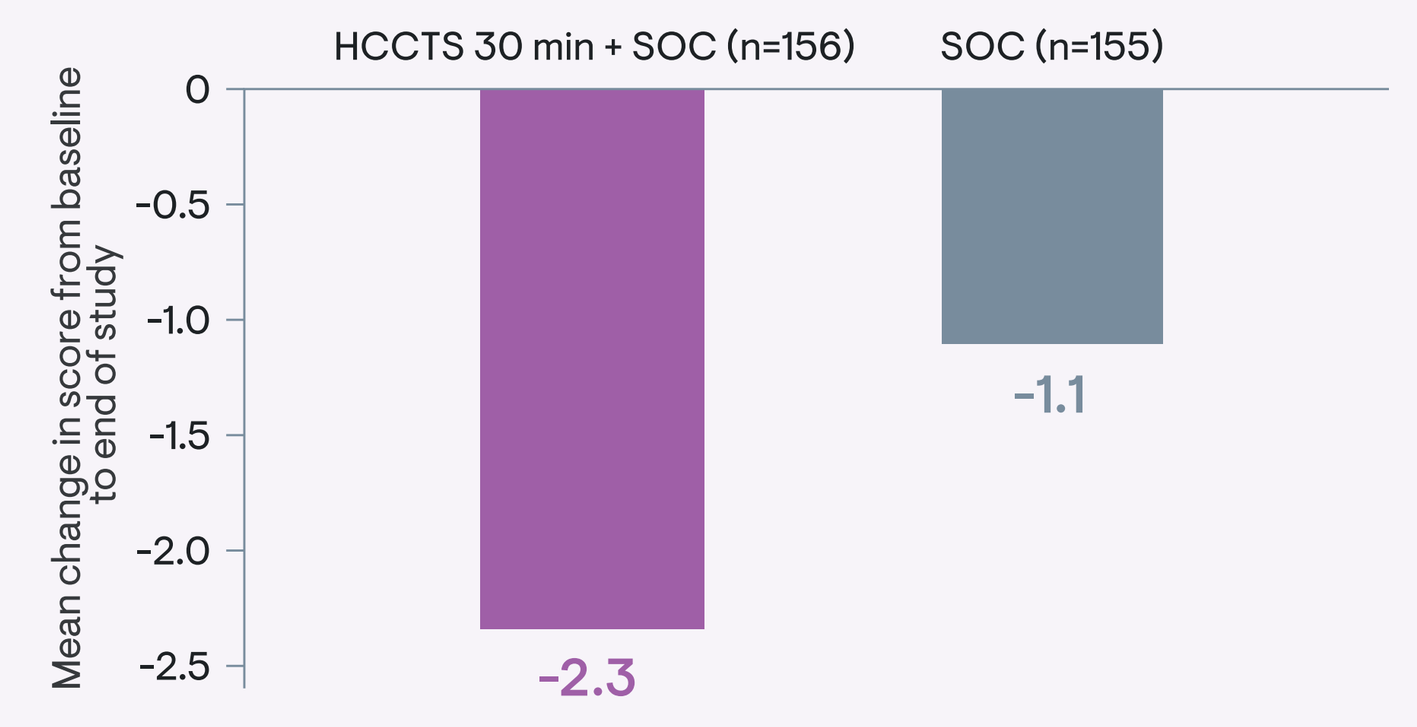
Mean change in score from baseline to end of study
HCCTS 30 min + SOC (n=156)
SOC (n=155)
0
-0.5
-1.0
-1.5
-2.0
-2.5
-2.3
-1.1

Mean score in mPDI-6 (sleep)

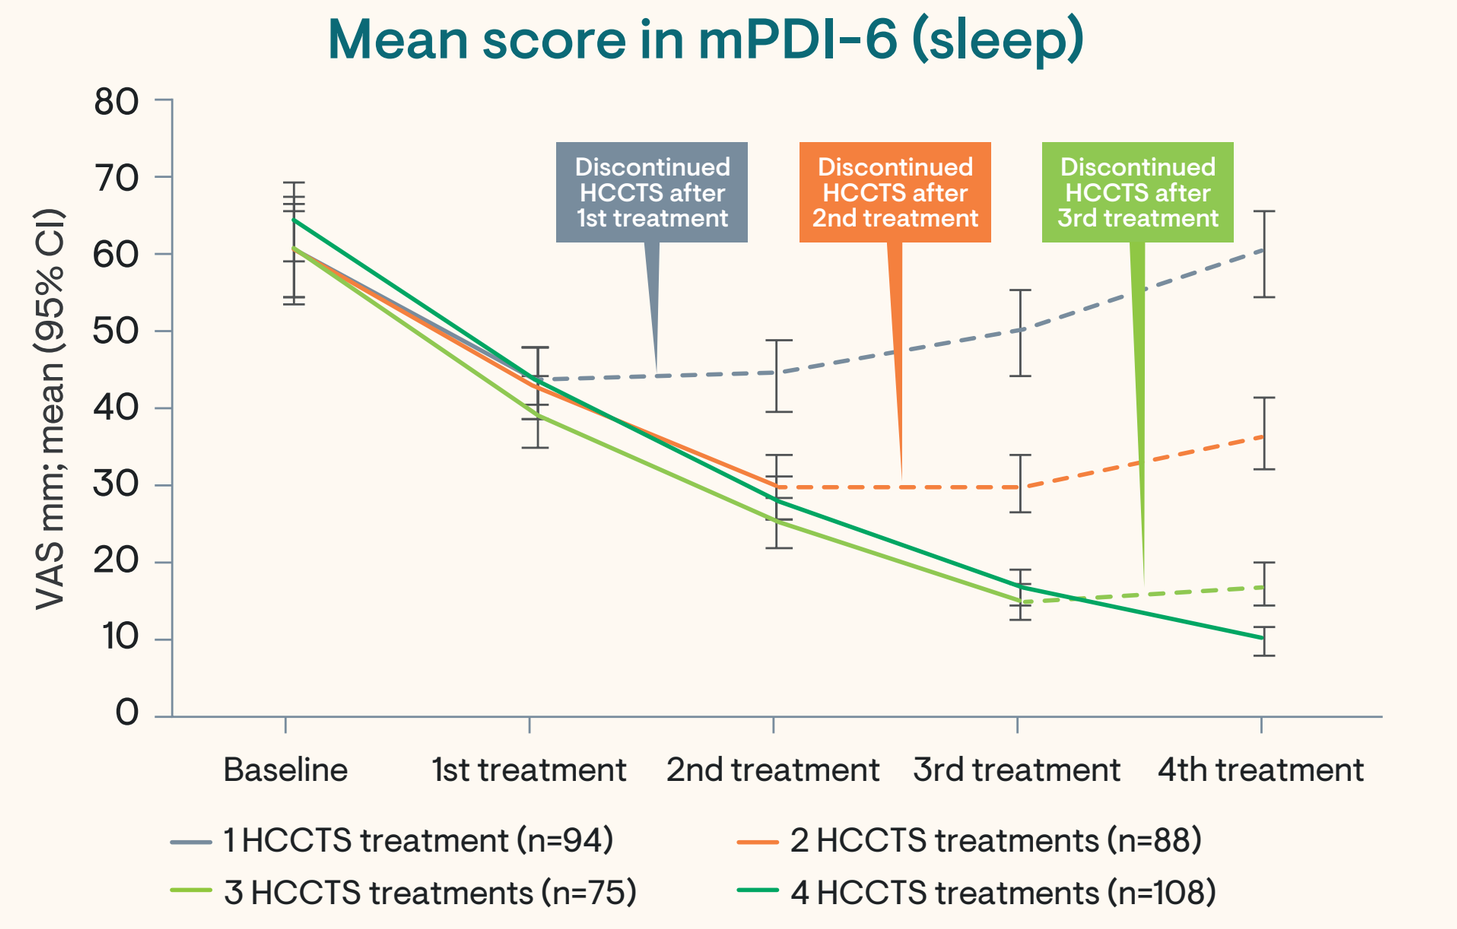
VAS mm; mean (95% CI)
80 70 60 50 40 30 20 10 0
Discontinued HCCTS after 1st treatment
Discontinued HCCTS after 2nd treatment
Discontinued HCCTS after 3rd treatment
Baseline
1st treatment
2nd treatment
3rd treatment
4th treatment
1 HCCTS treatment (n=94)
2 HCCTS treatments (n=88)
3 HCCTS treatments (n=75)
4 HCCTS treatments (n=108)

Application-Related Adverse Events

Most commonly reported AEs across PACE and CASPAR	Number (%) of patients experiencing AEs in group receiving HCCTS 30 min + SOC (regardless of number of treatments, n=156): Baseline - end of study	Number (%) of patients experiencing AEs with each successive treatment: 1st (n=365)	2nd (n=271)	3rd (n=183)	4th (n=108)
APPLICATION-SITE PAIN	44 (28.2)	240 (65.8)	129 (47.6)	68 (37.2)	30 (27.8)
BURNING SENSATION	14 (9.0)	57 (15.6)	22 (8.1)	24 (13.1)	5 (4.6)
APPLICATION-SITE ERYTHEMA	12 (7.7)	190 (52.1)	111 (41.0)	58 (31.7)	27 (25.0)

References

1. Gylfadottir SS, et al. Pain. 2020;161:574–583.
2. Yang H, et al. Front Endocrinol (Lausanne). 2019;10:929.
3. Feldman EL, et al. Nat Rev Dis Primers. 2019;5:42.
4. Eid SA, et al. Neuron. 2023;111:2623–2641.
5. Eastman DM, Dreyer MA. Atlas of the Diabetic Foot. 2022;74–90.
6. Vinik AI, et al. J Curr Med Res Opin. 2019;2:388–401.
7. Vinik AI, et al. BMC Neurol. 2016;16:251.
8. Überall MA, et al. Diabetes. 2024;73(Suppl 1):1892–LB.
9. Kim HS, et al. J Korean Med Sci. 2018;33:e213.
10. Flather M, et al. Clin Trials. 2006;3:508–512.
11. Averitas Pharma. Data on file.

Disclosures

Samuel Allen, PhD: Full-time employee of Averitas Pharma

Audrey Carnevale, PhD: Full-time employee of Averitas Pharma

Lizandra Marcondes, MD, PhD: Full-time employee of Averitas Pharma

Mariëlle Eerdekens, MD: Full-time employee of Grünenthal GmbH

Acknowledgments

Editorial and creative services provided by NexGen Healthcare Communications, funded by Averitas Pharma.

AE, adverse event; BL, baseline; BMI, body mass index; BPI-DN, Brief Pain Inventory – Diabetic Neuropathy; CI, confidence interval; DPN, diabetic peripheral neuropathy; HCCTS, high-concentration capsaicin 8% topical system; mPDI, modified Pain Disability Index; pDPN, painful diabetic peripheral neuropathy; QoL, quality of life; QoL-DN, Quality of Life – Diabetic Neuropathy; RCT, randomized controlled trial; RWE, real-world evidence; SOC, standard of care; VAS, visual analog scale.

This poster exhibit is sponsored by Averitas Pharma.