Ongoing treatment with high-concentration capsaicin 8% topical system results in progressive pain reduction for painful diabetic peripheral neuropathy: a retrospective real-world analysis

490-P

Gary Graf, APRN[1], Sam Allen, PhD[2], Audrey Carnevale, PhD[2]

[1]Cotton O'Neil Endocrinology and Diabetes Center, Topeka, KS, USA 66606, [2]Averitas Pharma, Morristown, NJ, USA 07960.

Background

- Painful diabetic peripheral neuropathy (PDPN) is a chronic complication of diabetes associated with reduced quality of life and significant morbidity[1,2]
- In PDPN, prolonged hyperglycemia leads to damage to peripheral nerves,[3] including C and Aδ nerve fibers that express transient receptor vanilloid potential 1 (TRPV1) receptors.[4] These receptors play a pivotal role in pain signaling.[4] Patients with diabetes have a reduced epidermal nerve fiber density (ENFD) compared with healthy controls, which can result in dysfunctional TRPV1-expressing fibers and debilitating pain often felt in the extremities[5,6]
- Progression of PDPN is linked to diabetic ulcers, infections, and, in cases of serious injury, foot amputation.[7] PDPN is further complicated by protective sensory loss and an increase in pain, and the disease will continue to progress if left untreated[7]
- Nerve damage is accelerated by poor microcirculation.[8] Glycemic control alone is insufficient to prevent the progression of PDPN and it is necessary to screen all patients with diabetes regularly for PDPN and initiate appropriate treatment in a timely fashion[8]
- In the USA, capsaicin 8% topical system (Figure 1) is approved by the FDA for the treatment of neuropathic pain associated with PDPN of the feet and post-herpetic neuralgia[9]
- Capsaicin is an agonist of TRPV1 receptors expressed on peripheral sensory nerve fibers, and selectively targets the TRPV1 receptors located on C and Aδ nerve fiber terminals.[4] Capsaicin 8% topical system induces reversible neurolysis of sensory nerve fiber terminals, resulting in relief from PDPN as early as 1 week after application (Figure 2). There is evidence that, following capsaicin 8% topical system treatment, the reduced ENFD in PDPN patients regenerates with a more "healthy" phenotype (less spontaneously active and with improved function)[10,11]
- Based on a small subset of biopsy data, the pain-relieving effects of a single application may not persist in the long term, owing to the chronic nature of diabetes and the progressive nature of DPN[10]
- In the PACE trial (NCT01478607), patients treated with repeated applications of 30-min (n=156) capsaicin 8% + standard of care (SOC) demonstrated continual reductions in average daily Numerical Pain Rating Scale (NPRS) scores compared with SOC alone over 12 months[12]
 - The percentage of patients achieving a clinically meaningful response (≥30% reduction from baseline in average daily pain) steadily increased with each subsequent application[12]

Purpose

- In order to better understand the effects of ongoing treatment and patients' response to capsaicin 8%, this retrospective, real-world, single-center case series explored whether patients receiving ongoing treatment with capsaicin 8% topical system for PDPN reported a progressive response in pain reduction, as assessed by NPRS values

Methods

- In our center, 18 patients had received ≥3 applications of capsaicin 8% topical system
- Of these 18 patients, 6 were not included in the final data set owing to missing NPRS scores
- Patients received ongoing treatment with capsaicin 8% topical system with approximately 12–16-week treatment intervals
- Efficacy, defined as a reduction in pain, was measured using the NPRS with values from 0 to 10, with 0 meaning "no pain" and 10 meaning "worst possible pain"
- The pre-application pain intensity score (NPRS) was captured by the clinician, as part of routine practice, prior to each administration of capsaicin 8% topical system
- The post-application pain intensity score (NPRS) was measured during an evaluation preceding the subsequent administration of capsaicin 8% topical system

Conclusions

- In a real-world setting, the majority of patients with PDPN achieved a clinically meaningful response to capsaicin 8% treatment, with significant reduction in average pain scores
- With each successive treatment, pain levels were further reduced. This is consistent with previous findings showing a progressive response in patients with PDPN[9,11]
- We hypothesize that this apparent progressive response is due to capsaicin-induced regeneration and restoration of more normal nerve fiber phenotypes. Other mechanisms that may be involved include gradual sensitization of the TRPV1 receptors, decreases in central sensitization, and/or increased patient activity, leading to beneficial effects on pain
- In PDPN, the peripheral nerves are under continued attack from hyperglycemia, leading to aberrant sensory nerve fiber terminals.[9] This persistent damage may underlie the return of pain and other changes commonly reported in patients with DPN
- As a chronic complication of diabetes, PDPN requires chronic treatment; in the case of capsaicin 8%, this involves timely and repeated re-application

Figure 1: Application of the capsaicin 8% topical system, which must be done by a HCP, in the office

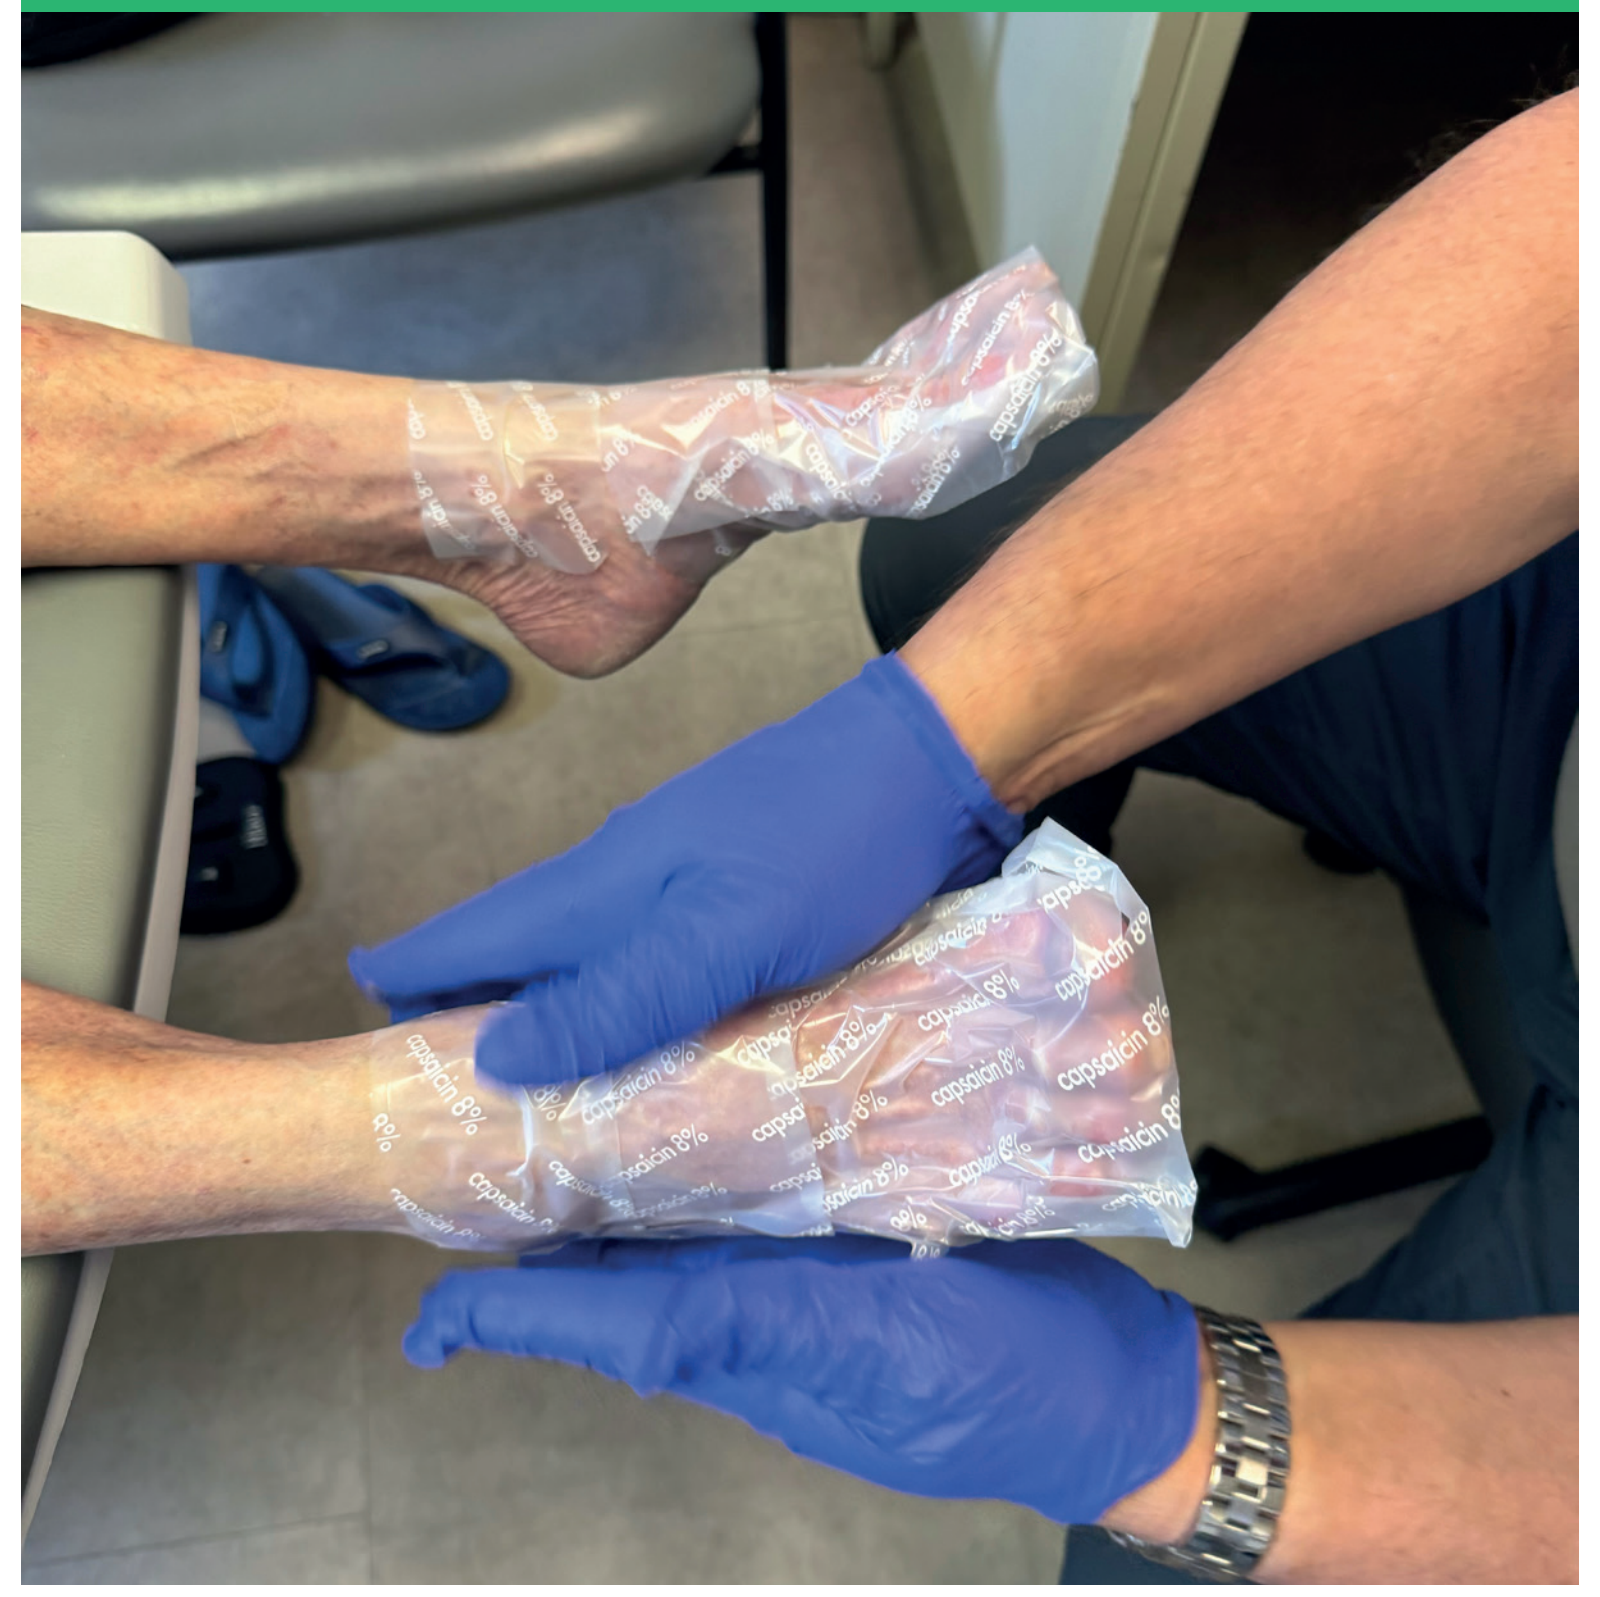

Figure 2: Hypothesized changes in nerve fiber anatomy following capsaicin 8% topical system in PDPN

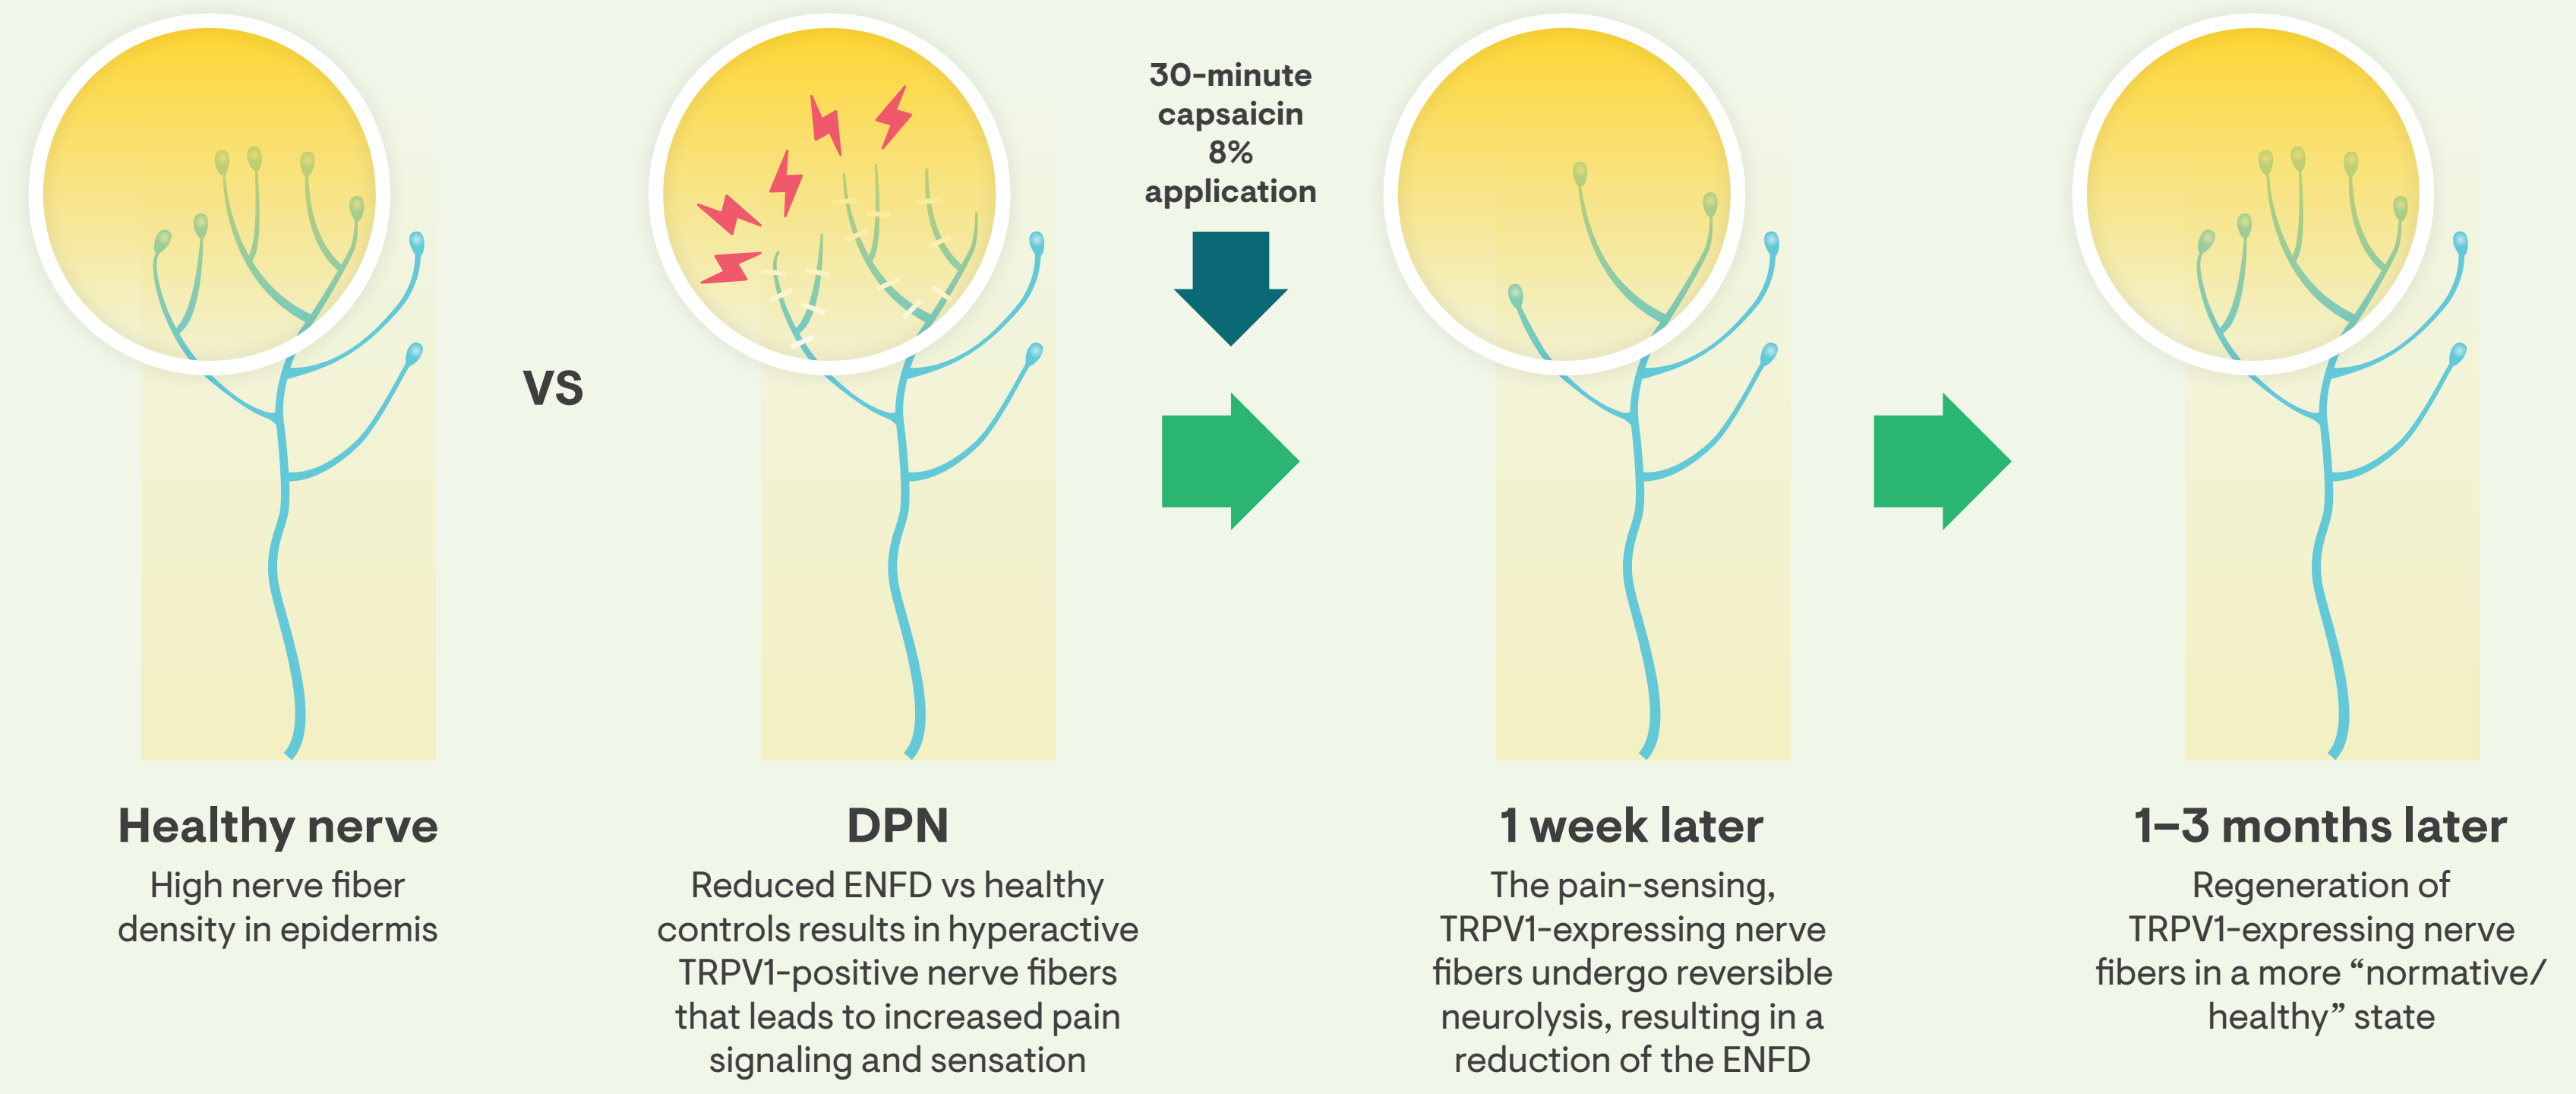
VS
30-minute capsaicin 8% application
Healthy nerve
High nerve fiber density in epidermis
DPN
Reduced ENFD vs healthy controls results in hyperactive TRPV1-positive nerve fibers that leads to increased pain signaling and sensation
1 week later
The pain-sensing, TRPV1-expressing nerve fibers undergo reversible neurolysis, resulting in a reduction of the ENFD
1–3 months later
Regeneration of TRPV1-expressing nerve fibers in a more "normative/ healthy" state

DPN, diabetic peripheral neuropathy; ENFD, epidermal nerve fiber density; PDPN, painful diabetic peripheral neuropathy; TRPV1, transient receptor vanilloid potential 1.

Figure 3. Effect of capsaicin 8% on pain intensity scores in patients receiving repeated treatments

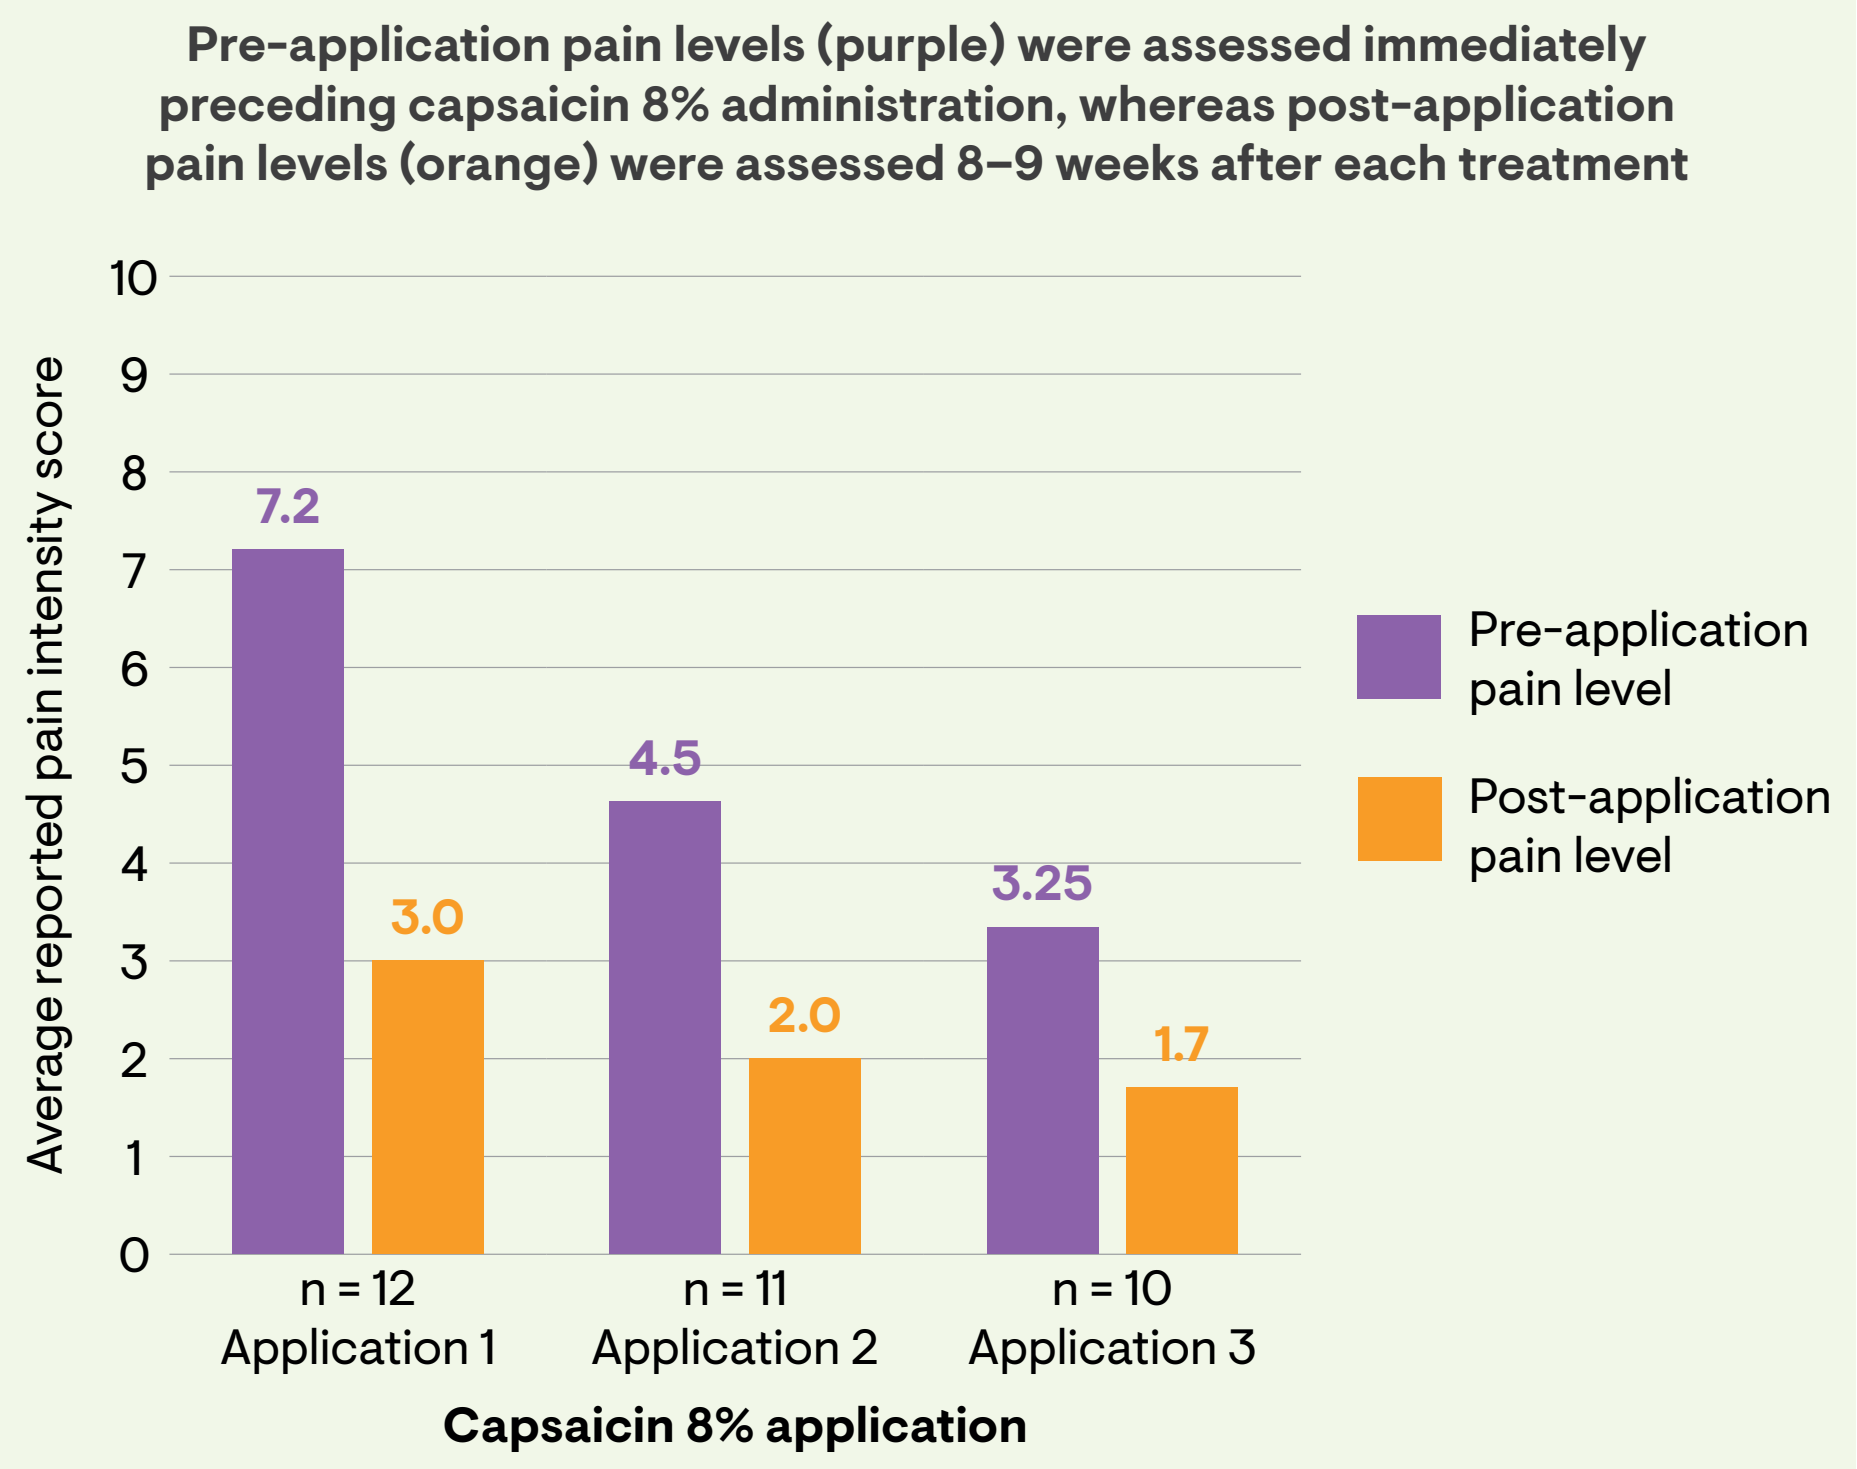
Pre-application pain levels (purple) were assessed immediately preceding capsaicin 8% administration, whereas post-application pain levels (orange) were assessed 8–9 weeks after each treatment
Average reported pain intensity score
10
9
8
7
6
5
4
3
2
1
0
7.2
3.0
4.5
2.0
3.25
1.7
n = 12
Application 1
n = 11
Application 2
n = 10
Application 3
Capsaicin 8% application
Pre-application pain level
Post-application pain level

Patient characteristics

- All patients reported an average NPRS score of ≥4 at baseline
- Eleven patients (61.1%) were male, and seven patients (38.9%) were female
- All patients had type 2 diabetes mellitus (T2DM)
 - The mean (standard deviation [SD]) duration of T2DM between diagnosis and treatment was 16.5 (7.70) years
 - The year of T2DM diagnosis was not available for three patients
- The mean (SD) age and body mass index were 66.6 (11.9) years and 33.4 (5.33) kg/m^2, respectively

Results

- The majority of patients (88.9%) reported a ≥50% reduction in pain from baseline on average in response to capsaicin 8% topical system throughout their treatments
 - Fifteen patients (83.3%) had the first response with the first treatment
 - One patient (5.6%) had the first response on the second treatment
 - Two patients (11.1%) did not respond to treatment
- The data show a progressive reduction in average pain intensity following each application with capsaicin 8% topical system (Figure 3)
 - After each treatment, the pain returned, albeit to lower levels than at baseline

Limitations

- This retrospective case series included a small number of patients from a single center and therefore the data cannot be generalized to larger populations

References

1. Gylfadottir SS, et al. Pain. 2020;161(3):574–83.
2. Yang H, et al. Front Endocrinol (Lausanne). 2019;10:929.
3. Feldman EL, et al. Nat Rev Dis Primers. 2019;5(1):42.
4. James CF, et al. Sisli Etfal Hastan Tip Bul. 2022;56(1):1–20.
5. Pittenger GL, et al. Diabetes Care. 2004;27(8):1974–9.
6. Facer P, et al. BMC Neurol. 2007;23:7:11.
7. Eastman DM & Dreyer DA. Neuropathic ulcer. In: StatPearls [Internet]. Treasure Island (FL): StatPearls Publishing; 2024 Jan-. Available from: https://www.ncbi.nlm.nih.gov/books/NBK559214/. Accessed April 2024.
8. Eid SA, et al. Neuron. 2023;111(17):2623–41.
9. QUTENZA® [prescribing information]. Morristown, NJ: Averitas Pharma, Inc.
10. Anand P, et al. Front Neurol. 2022;13:998904.
11. Kennedy WR, et al. J Pain. 2010;11(6):579–87.
12. Vinik AI, et al. J Curr Med Res Opin. 2019;2(12):388–401.

Disclosures

Gary Graf, APRN: Consultant to Averitas Pharma
Sam Allen, PhD: Full-time employee of Averitas Pharma
Audrey Carnevale, PhD: Full-time employee of Averitas Pharma

Acknowledgements

Editorial and creative services provided by NexGen Healthcare Communications, funded by Averitas Pharma.

Editorial and creative services provided by NexGen Healthcare Communications, funded by Averitas Pharma.